ReishiMax protects liver and improves liver functions in an experimental hepatitis model

Wen-Chuan Lin, Yue-Wen Wu, Ming-Chun Xie, Jia-Shi Zhu

Dept Pharmacology, College of Chinese Medicine, Taichung ROC; Pharmanex China Pharmacology Center, Beijing China; and Pharmanex, Provo, UT

Abstract

Literature reported that Reishi (*Ganoderma lucidum*) is capable of protecting liver. This study is to examine the liver protective functions of ReishiMax (RM), a proprietary product containing both extract of *G. lucidum* fruit body and cracked spores of *G. lucidum* (Pharmanex). We used a CCl_4 (20%, 0.5ml/rat)-induced liver injury rat model. RM was given daily to rats at a dose of 208, 624, or 1664 mg/kg by gavage, starting one week prior to CCl_4 injection, and continuously after CCl_4 injection for 8 weeks to the end of experiment. CCl_4 injection caused liver injuries in the vehicle control group, featuring increase in serum GOT and GPT, liver collagen, and spleen weight, decreases in serum albumin and liver total protein. RM supplementation reduced the transaminases (p<0.05~0.01) and liver collagen (p<0.01); prevented the reductions of serum albumin (p<0.05) and liver total protein (p<0.05); reduced spleen weight (p<0.05). Histopathology examination showed apparent improvement of liver structure in RM-treated rats. Our results demonstrated that RM protects the liver from chemical injuries, improves liver functions, and prevents injury-associated liver fibrosis/cirrhosis.

Introduction

Ganoderma lucidum (Reishi)

- Used for millenium as a longevity-promoting herbal tonic
- In animal studies, enhancing immune functions, improving glucose metabolism, liver protection, anti-cancer, antioxidant, and benefiting nerve system
- Active components:
 (1) *G. lucidum* polysaccharides
 (2) Highly oxygenated triterpenes
- *G. lucidum* triterpenes'functions:
 (1) actinociceptive,
 (2) anti-HIV,
 (3) inhibition of release of histamine,
 (4) inhibition of HMG-CoA reductase,
 (5) inhibition of angiotensin convertin enzyme,
 (6) antihepatoma, antihepatotoxic, and improving liver functions

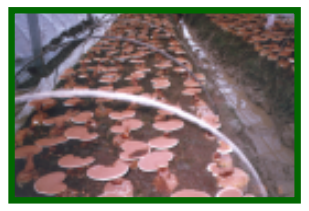

Ganoderma lucidum

Proprietary Manufacturing Process

Reishi extract

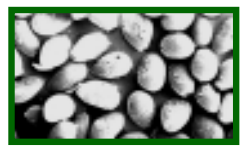

Reishi spores

Cracking Process

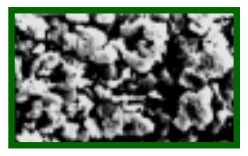

Cracked Reishi spores

ReishiMax

6.0% Reishi triterpenes

13.5% Reishi polysaccharides

To examine the liver protecting effects of ReishiMax in mice with CCl4-induced experimental hepatitis

Normal mice placebo control		(n=10)
Placebo		(n=12)
ReishiMax	208 mg/kg	(n=12)
ReishiMax	624 mg/kg	(n=12)
ReishiMax	1164 mg/kg	(n=12)
Silymarin	200 mg/kg	(n=12)

-1 0 1 2 3 4 5 6 7 8 (Weeks)

CCl4 100 mg p.o. twice every week

Reduction of serum transaminase (ALT)

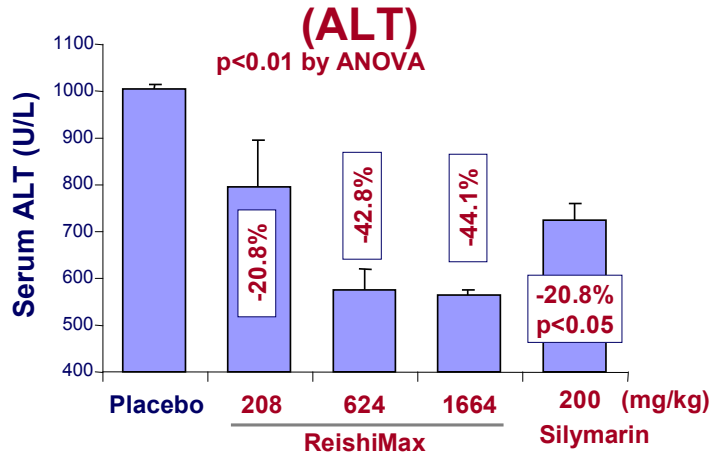

p<0.01 by ANOVA
Serum ALT (U/L)
1100
1000
900
800
700
600
500
400
-20.8%
-42.8%
-44.1%
-20.8%
p<0.05
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin

Reduction of Liver Hydroxyproline (collagen)

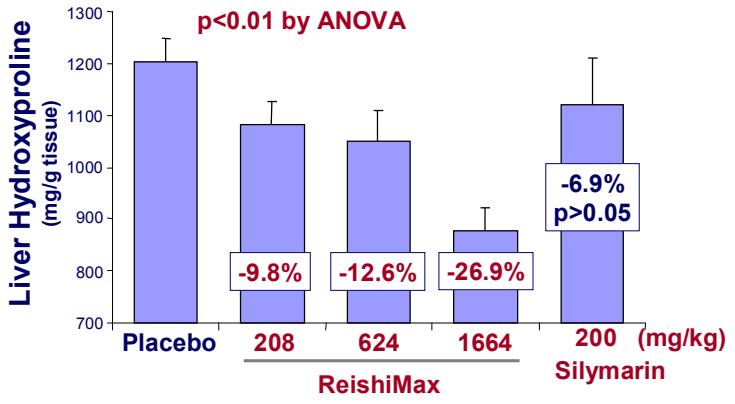

p<0.01 by ANOVA
Liver Hydroxyproline
(mg/g tissue)
1300
1200
1100
1000
900
800
700
-9.8%
-12.6%
-26.9%
-6.9%
p>0.05
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin

Liver architecture (HE staining)

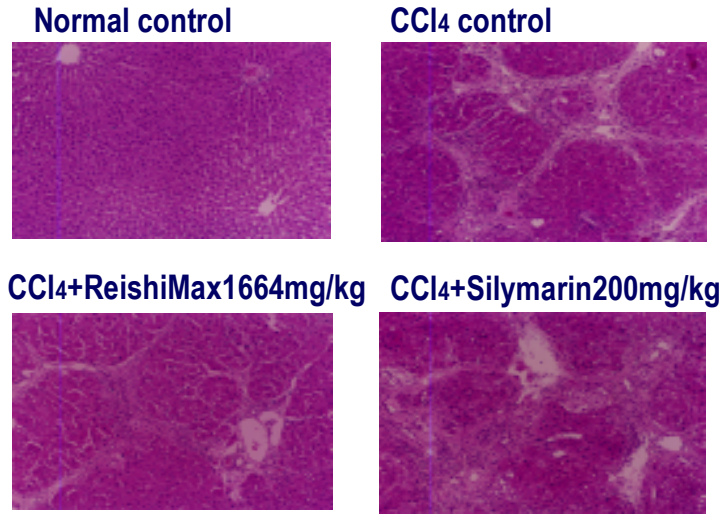

Normal control
CCl4 control
CCl4+ReishiMax1664mg/kg
CCl4+Silymarin200mg/kg

Increase in plasma albumin

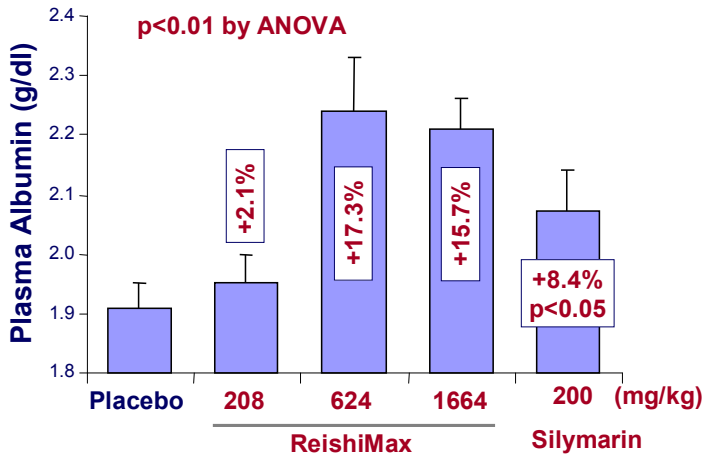

p<0.01 by ANOVA
Plasma Albumin (g/dl)
2.4
2.3
2.2
2.1
2.0
1.9
1.8
+2.1%
+17.3%
+15.7%
+8.4%
p<0.05
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin

Reduction of Spleen Weight

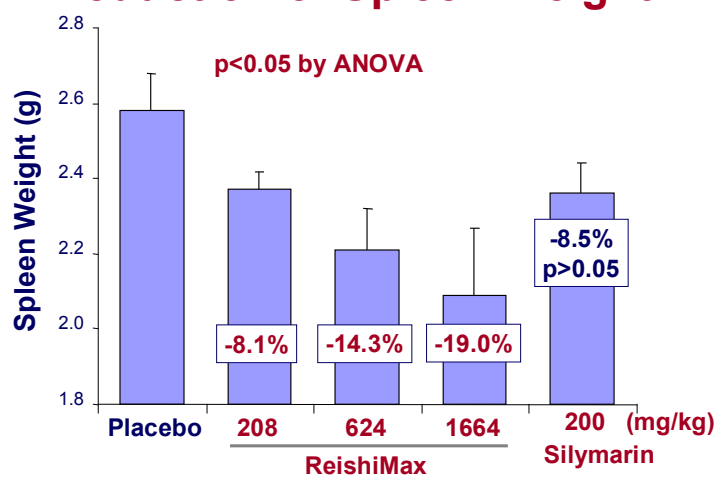

p<0.05 by ANOVA
Spleen Weight (g)
2.8
2.6
2.4
2.2
2.0
1.8
-8.1%
-14.3%
-19.0%
-8.5%
p>0.05
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin

Collagen content & structure (Masson's staining)

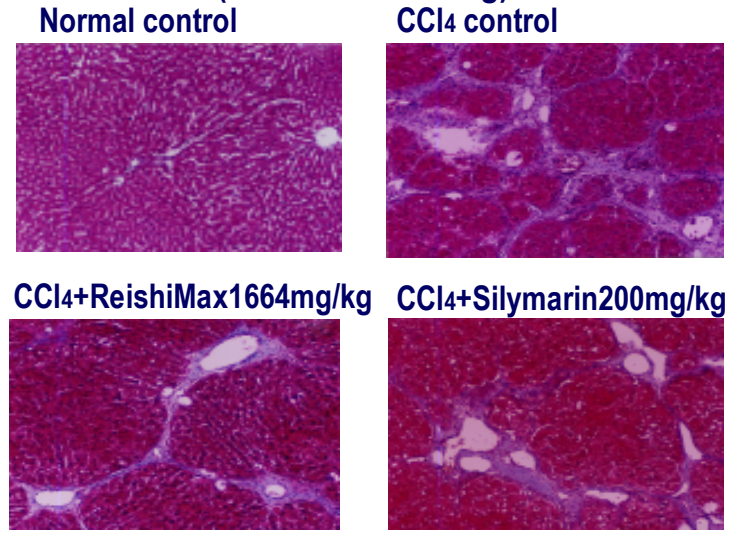

Normal control
CCl4 control
CCl4+ReishiMax1664mg/kg
CCl4+Silymarin200mg/kg

Increase in Liver Total Protein

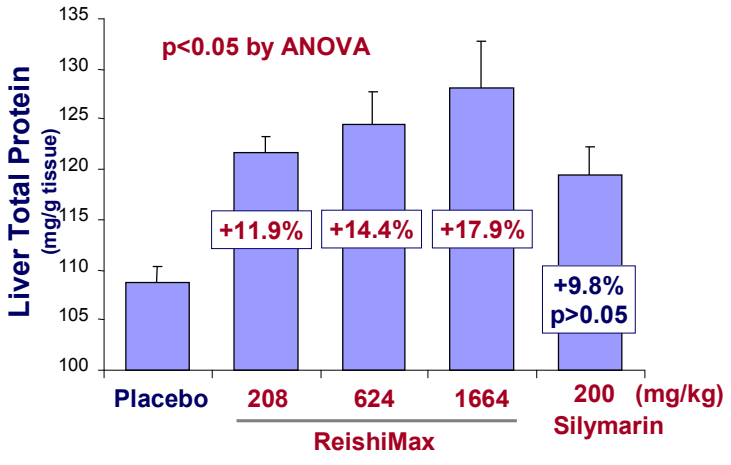

p<0.05 by ANOVA
Liver Total Protein
(mg/g tissue)
135
130
125
120
115
110
105
100
+11.9%
+14.4%
+17.9%
+9.8%
p>0.05
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin

Reduction of Severity of Fibrosis

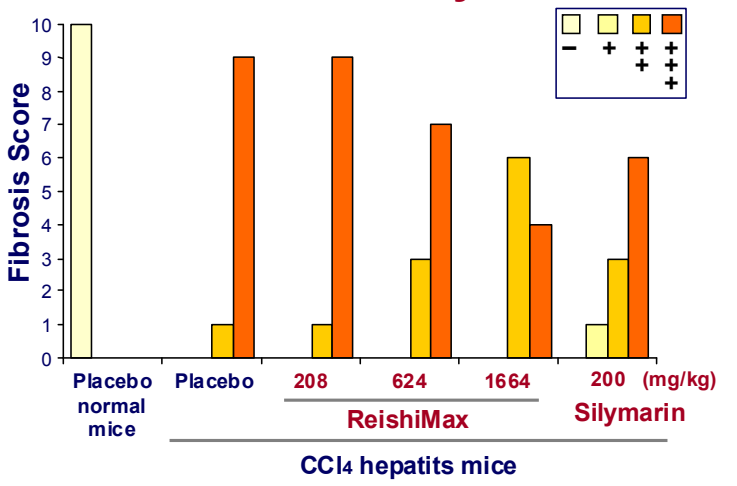

Fibrosis Score
10
9
8
7
6
5
4
3
2
1
0
−
+
++
+++
Placebo normal mice
Placebo
208
624
1664
200 (mg/kg)
ReishiMax
Silymarin
CCl4 hepatits mice

Summary

In mice with CCl4-induced experimental hepatitis, oral ReishiMax supplementation

↓Serum transaminases (ALT &AST)

↑Plasma albumin and total liver proteins

↓Liver collagen ↓Spleen weight

↓Severity of liver necrosis & fibrosis

Conclusion

Oral administration of ReishiMax protects the liver from chemical injuries, improves liver functions, and prevents injury-associated liver fibrosis/cirrhosis.